Designer gelators for controlled crystallisation of pharmaceuticals

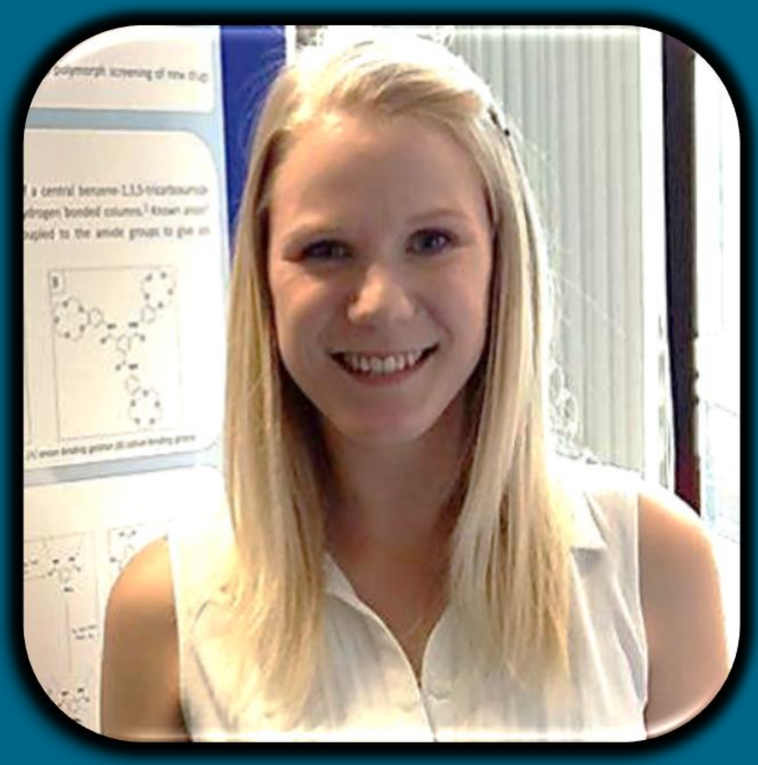

***Jessica L. Andrews*[A], *Jonathan W. Steed*[A], *Katharina Edkins*[B], *David J. Berry*[B] *and Jim McCabe*[C]**

jessica.andrews@durham.ac.uk

A: Department of Chemistry, Durham University, South Road, Durham, DH1 3LE, B: Department of Pharmacy, Durham University, Queen's Campus, Stockton, TS17 6BH , C: Astra Zeneca, Charter Way, Macclesfield, SK10 2NA

Introduction

Control of solid form is a key area of research in the pharmaceutical industry, owing to the differing physical properties of a drug's polymorphs. Solid form selective crystallisation can be achieved using a gel because the supramolecular network limits convection, controls nucleation and allows diffusion-limited crystal growth. For a gel network to control the outcome of a crystallisation, the gel fibres and drug molecules must interact. This has previously been achieved by mimicking a structural feature of a particular drug in the gelator.[1,2] This work targets a more 'general purpose' gelator, equipped with peripheral ion binding groups to interact with the counterion of pharmaceutical salts, tethering the drug to the gel fibre, promoting nucleation and favouring formation of one solid form over the others, without being specific to one particular drug.

Gel-Phase Crystallisation

Two different approaches to gel-phase crystallisation of will be compared to investigate the influence of the gel network on crystallisation.

ION BINDING

Ion binding gelators tether the drug to the gel fibre

The target gelators are composed of a central tris-amide or tris-urea moiety which self-assembles into hydrogen bonded columns. Known anion and cation binding units are coupled to the outer edge to trap the counterion of a drug, tethering the molecule to the gel fibre and providing a nucleation site. This function is not specific to the structure of one drug and is a first attempt at a 'general purpose' gelator for solid form control.

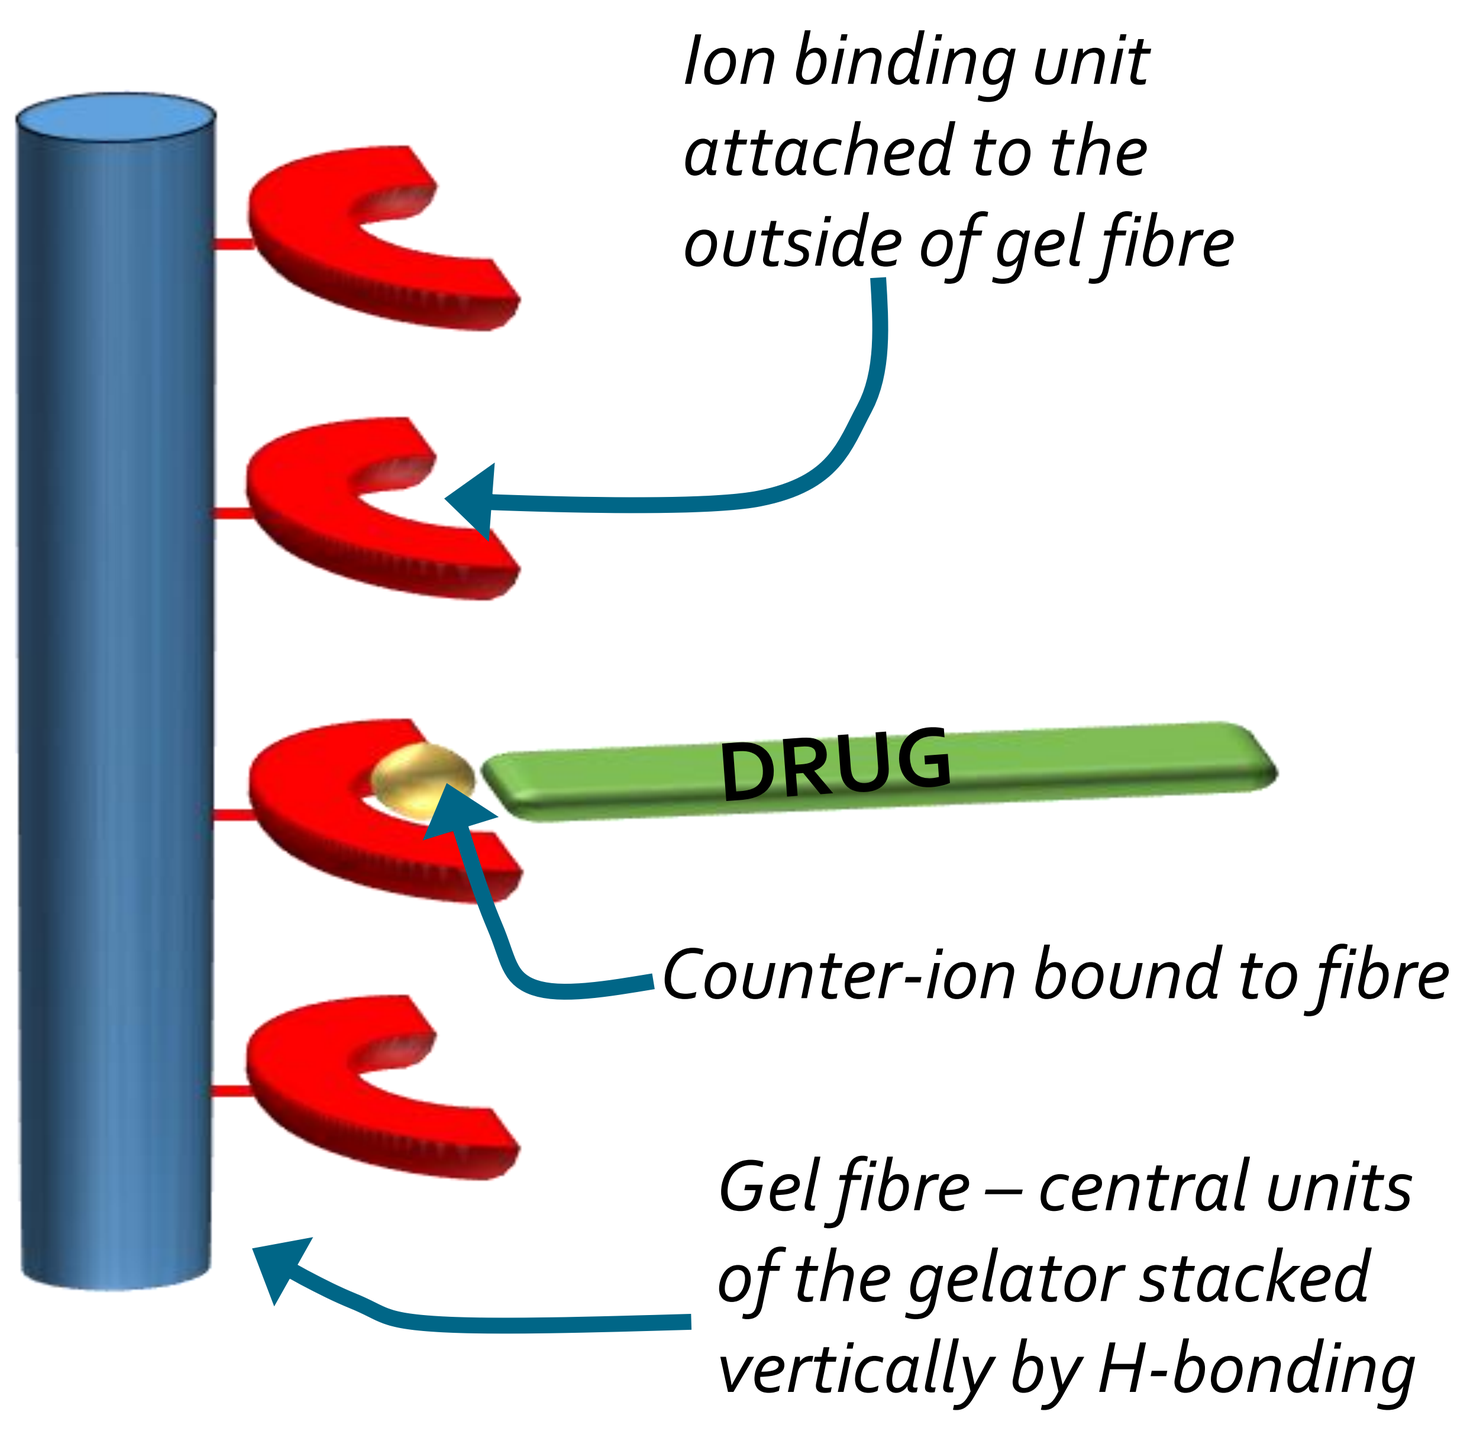

Ion binding unit attached to the outside of gel fibre
DRUG
Counter-ion bound to fibre
Gel fibre – central units of the gelator stacked vertically by H-bonding

Target Molecules

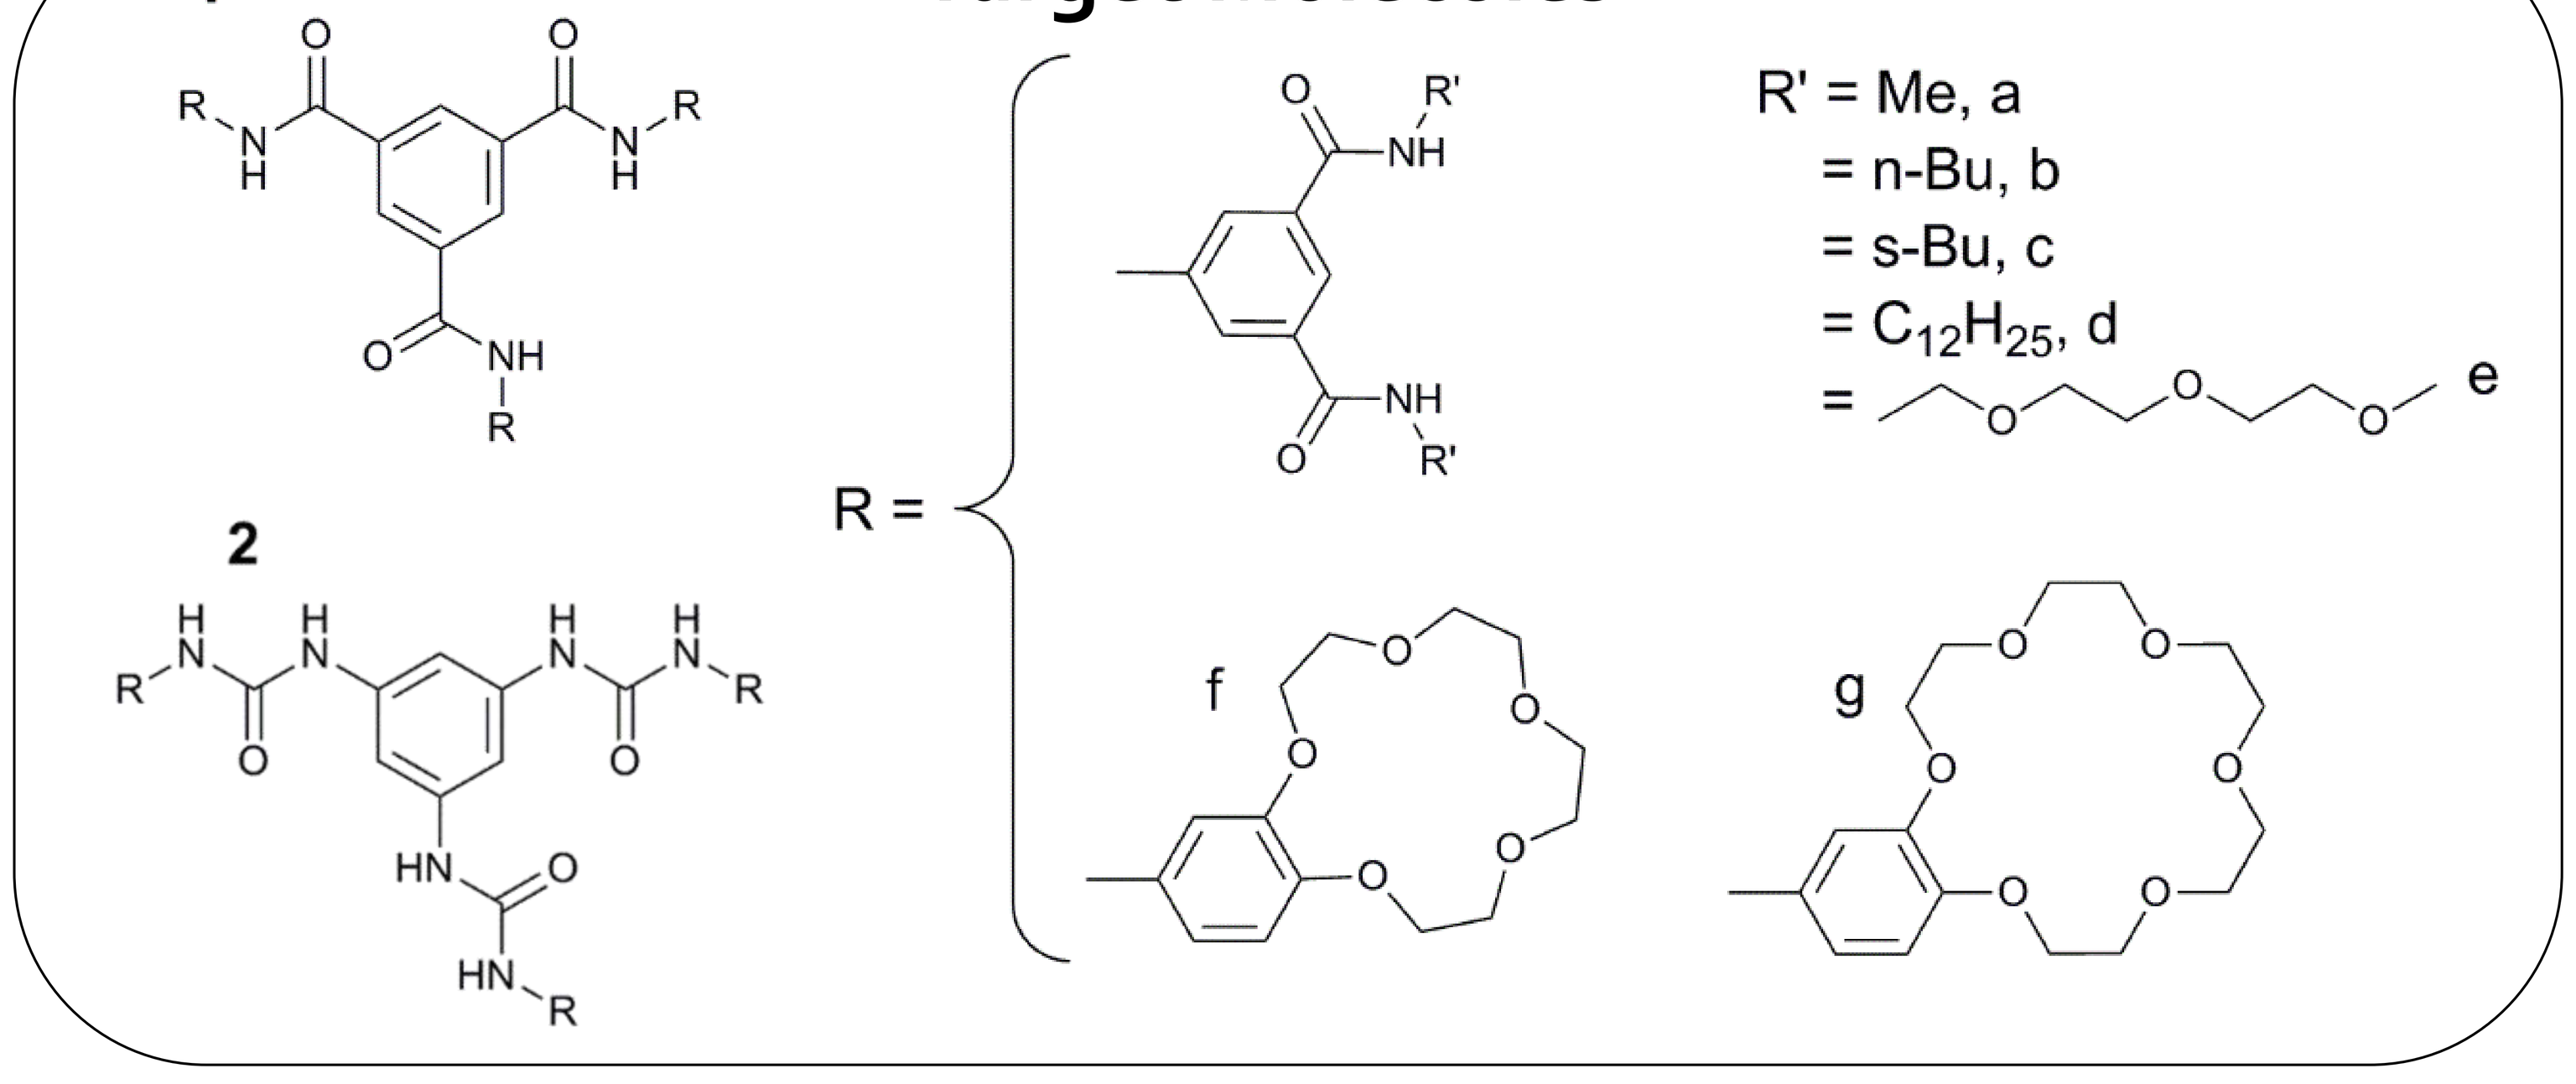

1
2
R =
R' = Me, a
= n-Bu, b
= s-Bu, c
= C12H25, d
e
f
g

Compound 1b

Solvent	Result at 20 mg/mL
Toluene	**G**
DCM	I
THF	S (gradually crystallises)
Chlorofom	**PG**
Ethyl Acetate	I
1,4-Dioxane	I
Acetone	I
Butan-2-one	I
Acetonitrile	I
Nitro Methane	I
n-Pentanol	S
1-Butanol	S
2-Butanol	S
1-Propanol	S
2-Propanol	S
Ethanol	S
Methanol	S
DMF	S
DMSO	S

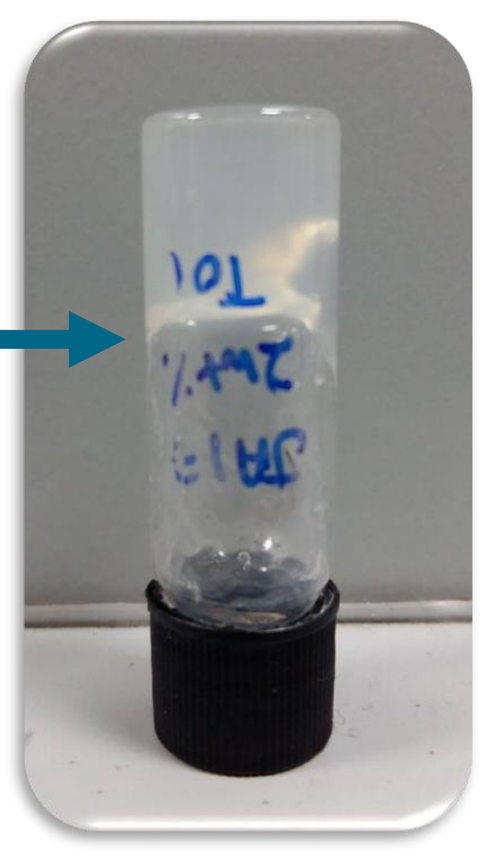

Preliminary gelation testing of amide linked gelators shows some success. However the increased H-bonding ability of ureas is likely to make these analogues better gelators.

G = Gel, PG = Partial Gel, S = Solution, I = Insoluble

Compound 1f

Solvent	Result at 20 mg/mL
Diethyl ether	I
Toluene	I
THF	**G (unstable, crystallises rapidly)**
DCM	S (gradually crystallises)
1,4-Dioxane	S (gradually crystallises)

DRUG MIMICKING

Drug mimicking gelators encourage templating

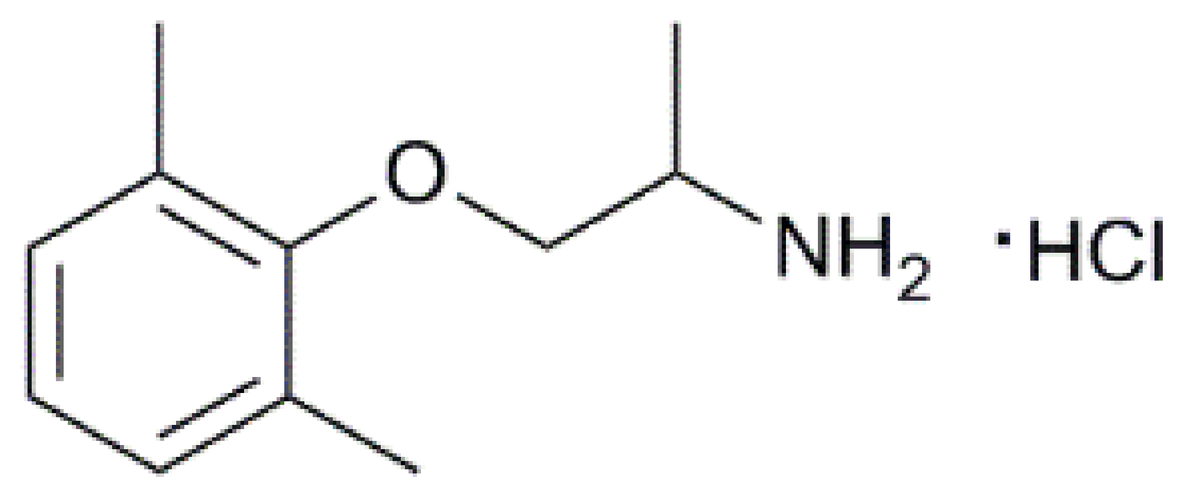

O
NH2
·HCl

The target drug is mexiletine hydrochloride. It's simple structure can easily be recreated in a gelator and it has six known polymorphs; offering large scope for solid form control.

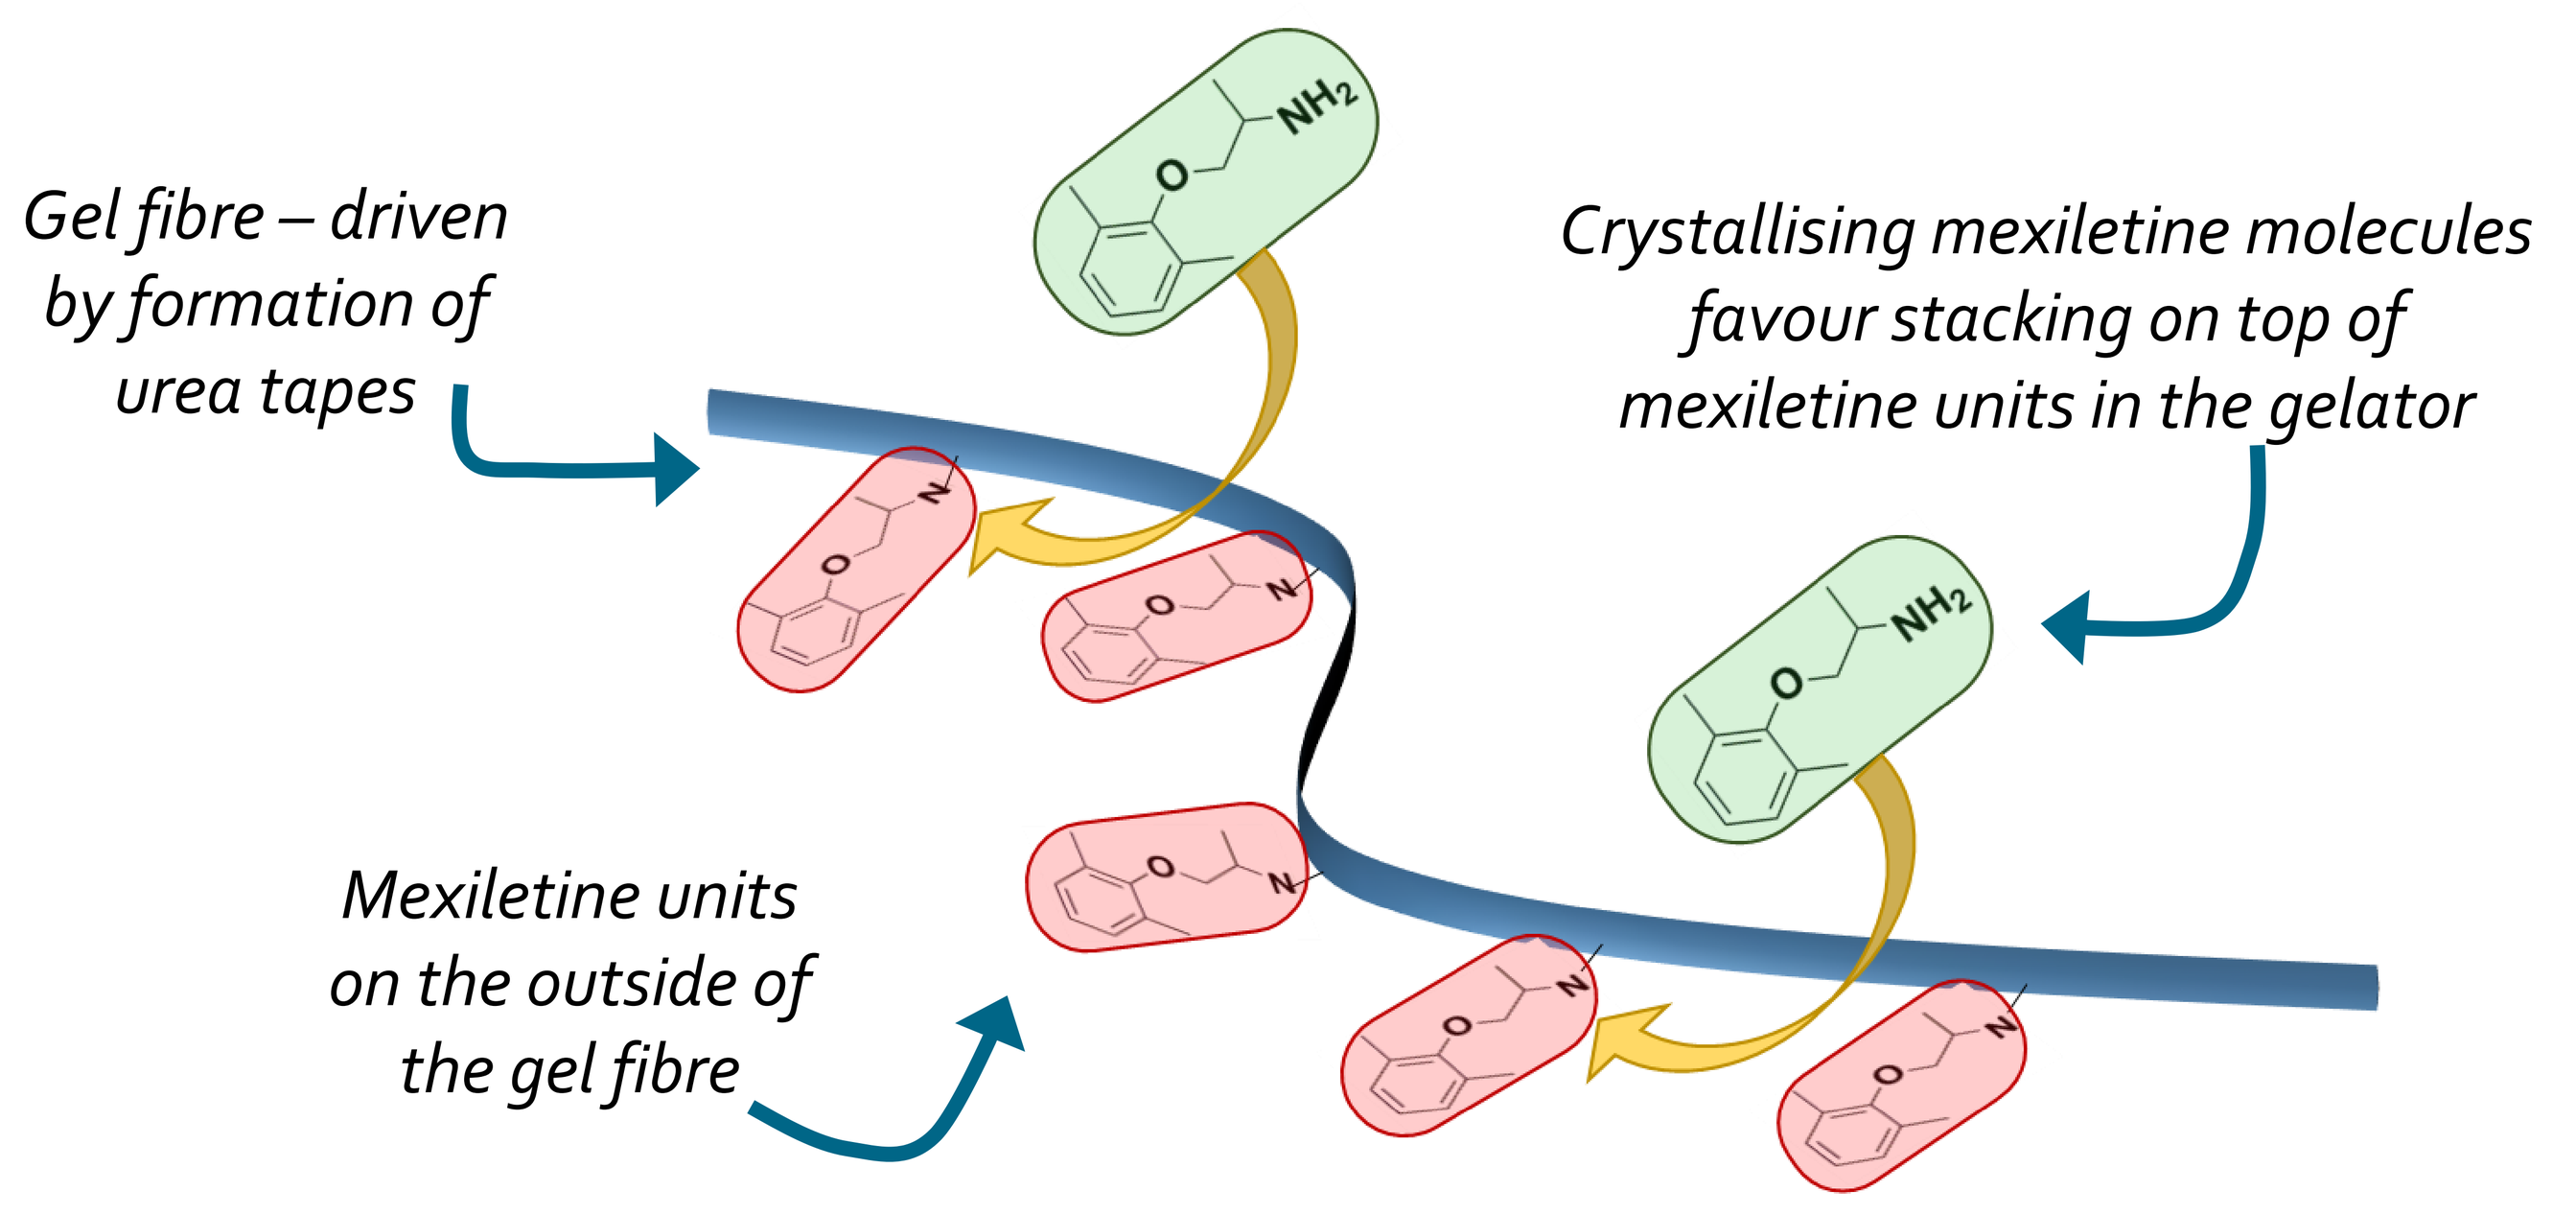

Gel fibre – driven by formation of urea tapes
Crystallising mexiletine molecules favour stacking on top of mexiletine units in the gelator
Mexiletine units on the outside of the gel fibre

The target gelators are analogues of the ion binding compounds with tris-amide or tris-urea units terminated by mexiletine. A bis-urea analogue is also proposed, to build off previous work in our group on polymorph control of ROY using analogous bis-urea gelators.[1] Whichever gelator is used, the self-assembled gel fibre will has pendant mexiletine units along its length. Crystallising mexiletine molecules stack on top of the ones on the fibre, due to π-stacking and hydrogen bonding, creating a templating effect.

Target Molecules

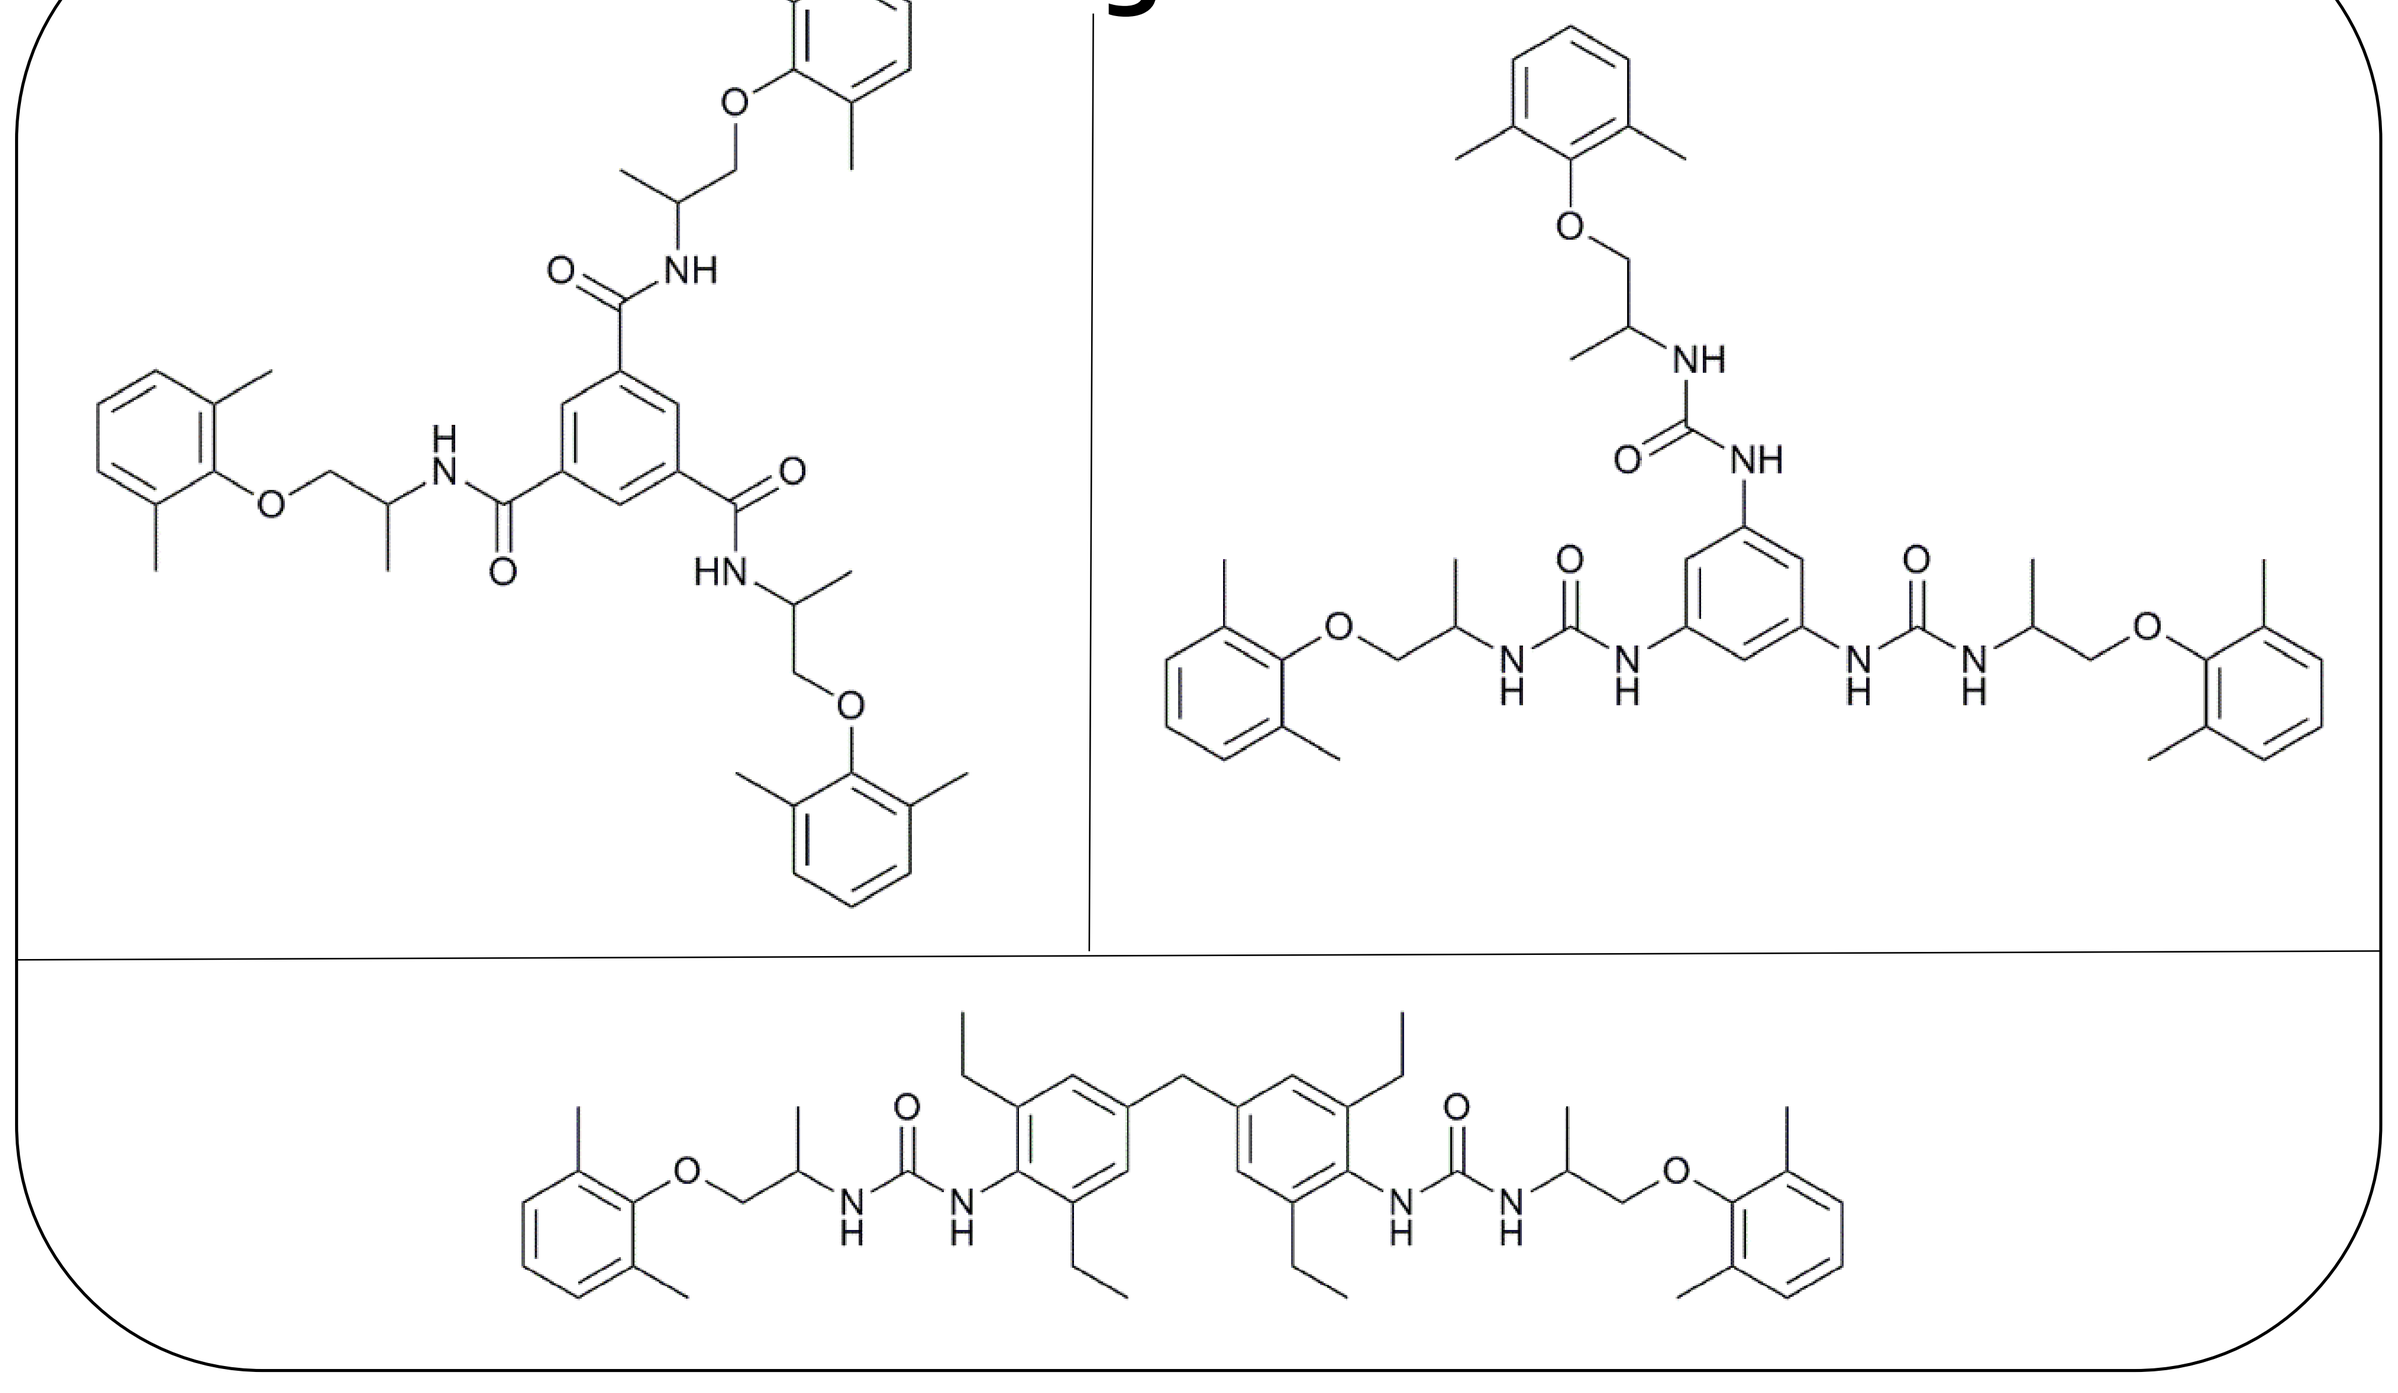

Conclusions & Future Work

- Urea and amide based supramolecular gelators have been designed and synthesis is underway.
- Preliminary results show gelation for two amide linked analogues.
- Mexiletine hydrochloride will be crystallised within the resulting gels to investigate the effect on solid form.
- The drug mimicking gelators will be compared to the 'general purpose' ion binding gelators to determine which is the more efficient strategy.

References and Acknowledgements

1. Foster J. A., Damodaran K. K., Maurin A., Day G. M., Thompson H. P. G., Cameron G. J., Cuesta Bernal J., Steed J. W., *Chem. Sci.*, 2017, **8**, 78-84.
2. Dawn A., Andrew K. S., Yufit D. S, Hong Y., Reddy J. P., Jones C. D., Aguilar J. A., Steed J. W., *Cryst. Growth. Des.*, 2015, **15**, 4591-4599.